LUDWIG-MAXIMILIANS-UNIVERSITÄT MÜNCHEN

FACULTY OF VETERINARY MEDICINE
CENTRE FOR CLINICAL VETERINARY MEDICINE

CLINIC FOR SWINE

Results from different diagnostic approaches concerning *Leptospira* spp. in terms of SMEDI litters

EDDICKS MATTHIAS [1], STRUTZBERG-MINDER KATRIN [2], EDDICKS LINA [3], SCHAACK NEELE [1], SEIFERT ANNIKA [1], TABELING ROBERT [4], RITZMANN MATHIAS [1]

[1] Clinic for Swine at the Centre for Clinical Veterinary Medicine, Ludwig-Maximilians-Universität München, Oberschleissheim, Germany, [2] IVD GmbH, Innovative Veterinary Diagnostics, Consultancy Laboratory of the German Veterinary Medical Society (GVMS) for *Leptospira* spp., Seelze, Germany, [3] Institute of Veterinary Pathology at the Centre for Clinical Veterinary Medicine, Ludwig-Maximilians-Universität, München, Germany, [4] Intervet Deutschland GmbH; MSD Animal Health, Unterschleissheim, Germany

DISCUSSION AND CONCLUSION

Kidney displayed the most suitable tissue for *Leptospira* spp. detection in SMEDI fetuses by qPCR. Heart blood of fetuses was often hemolytic and therefore not suitable for the serological diagnostics of *Leptospira* spp.. Highest titers against serovars Pomona and Bratislava were present in sera of sows from *Leptospira* spp. PCR positive litters.

BACKGROUND AND OBJECTIVES

Diagnostic material for PCR in terms of *Leptospira* ssp. associated abortion or stillbirth may include lung, liver, kidney or stomach content from affected fetuses or the detection of antibodies against *Leptospira* spp. in stillborn fetuses, if infected after the 70th day of gestation. In the present study we used different specimens for direct *Leptospira* spp. or MAT for antibody detection to evaluate adequate diagnostic approaches for *Leptospira* diagnostic in SMEDI cases.

MATERIALS AND METHODS

A total of 142 fetuses from 36 SMEDI litters from 16 farms were included in the present study. The inclusion criterion was the availability of serum samples from the corresponding dams. Serological examinations (MAT) of sow sera, fetal heart blood and PCR (1,2) examinations of fetal tissue pools (Kidney, Liver), stomach content and meconium from affected fetuses were carried out.

RESULTS

Antibodies against *Leptospira* spp. were present in 62.5 % of the farms and 44.4 % of the individual sow sera. Highest titers against serovars Pomona and Bratislava were present in sera of sows from *Leptospira* spp. PCR positive litters (Figure 2). Due to hemolysis, only 23.5 % heart blood samples of the fetuses could be examined by MAT; all revealed negative results. In total 14.1 % of the tissue pools were positive for *Leptospira* spp. by qPCR. Single sample examination revealed a detection rate of 95 % for kidney, 85 % for liver, 70 % for meconium and 65 % for stomach content (Table 1, Figure 1).

REFERENCES

1. Perez, L.J., Lanka, S., DeShambo, V.J., Fredrickson, R.L., Maddox, C.W., 2020. A Validated Multiplex Real-Time PCR Assay for the Diagnosis of Infectious Leptospira spp.: A Novel Assay for the Detection and Differentiation of Strains From Both Pathogenic Groups I and II. Frontiers in Microbiology 11, 457

2. Ferreira, A.S., Costa, P., Rocha, T., Amaro, A., Vieira, M.L., Ahmed, A., Thompson, G., Hartskeerl, R.A., Inacio, J., 2014. Direct detection and differentiation of pathogenic Leptospira species using a multi-gene targeted real time PCR approach. PLoS One 9, e112312.

Table 1: Overall positive sample pools (n = 142) and percentage of *Leptospira* spp. DNA positive tissues of all positive pools (n = 20) by PCR and total

	Pools	Kidney	Liver	Stomach content	Meconium
LipL32Gene	14.1 % (20/142)	95.0 % (19/20)	85.0 % (17/20)	65.0 % (13/20)	70.0 % (14/20)
16SGene	12.7 % (18/142)	90.0 % (18/20)	80.0 % (16/20)	70.0 % (14/20)	55.0 % (11/20)
Total	14.1 % (20/142)	95.0 % (19/20)	85.0 % (17/20)	80.0 % (16/20)	75.0 % (15/20)

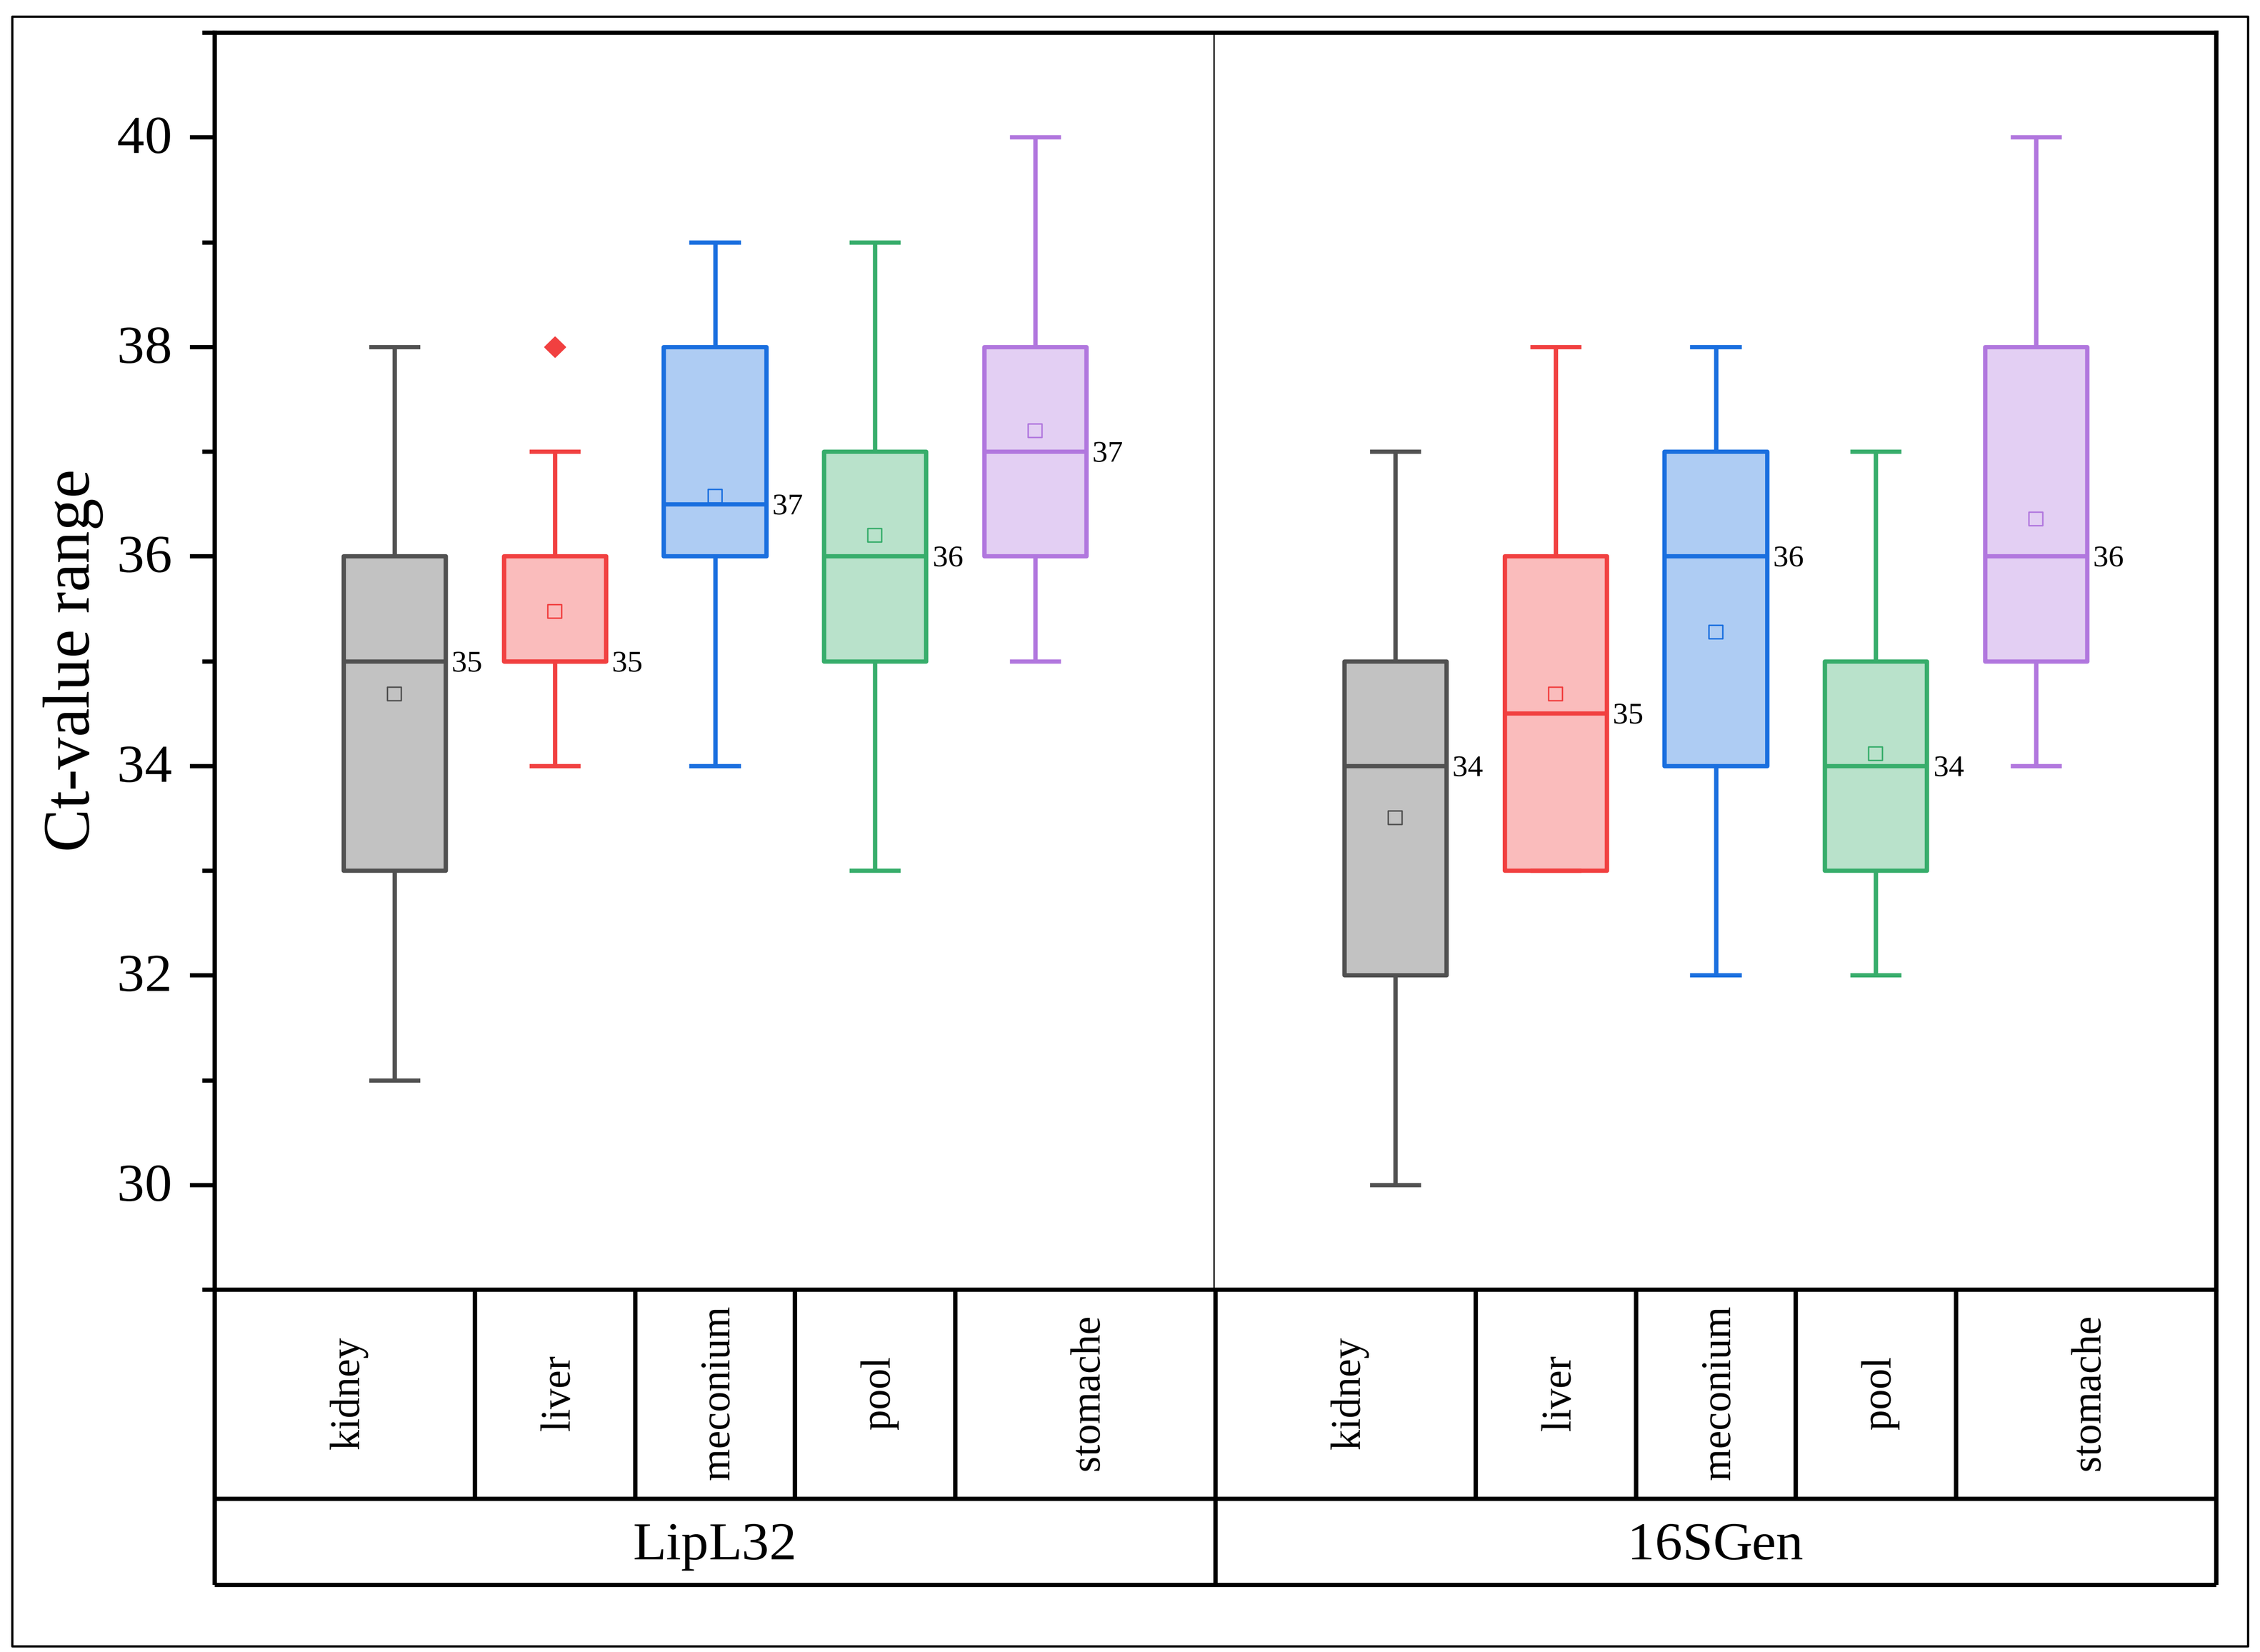

Ct-value range
40
38
36
34
32
30
35
35
37
36
37
34
35
36
34
36
kidney
liver
meconium
pool
stomache
LipL32
16SGen

Figure 1: Box of Ct-values including the median value from different tissues in the LipL32 PCR or 16SGene PCR

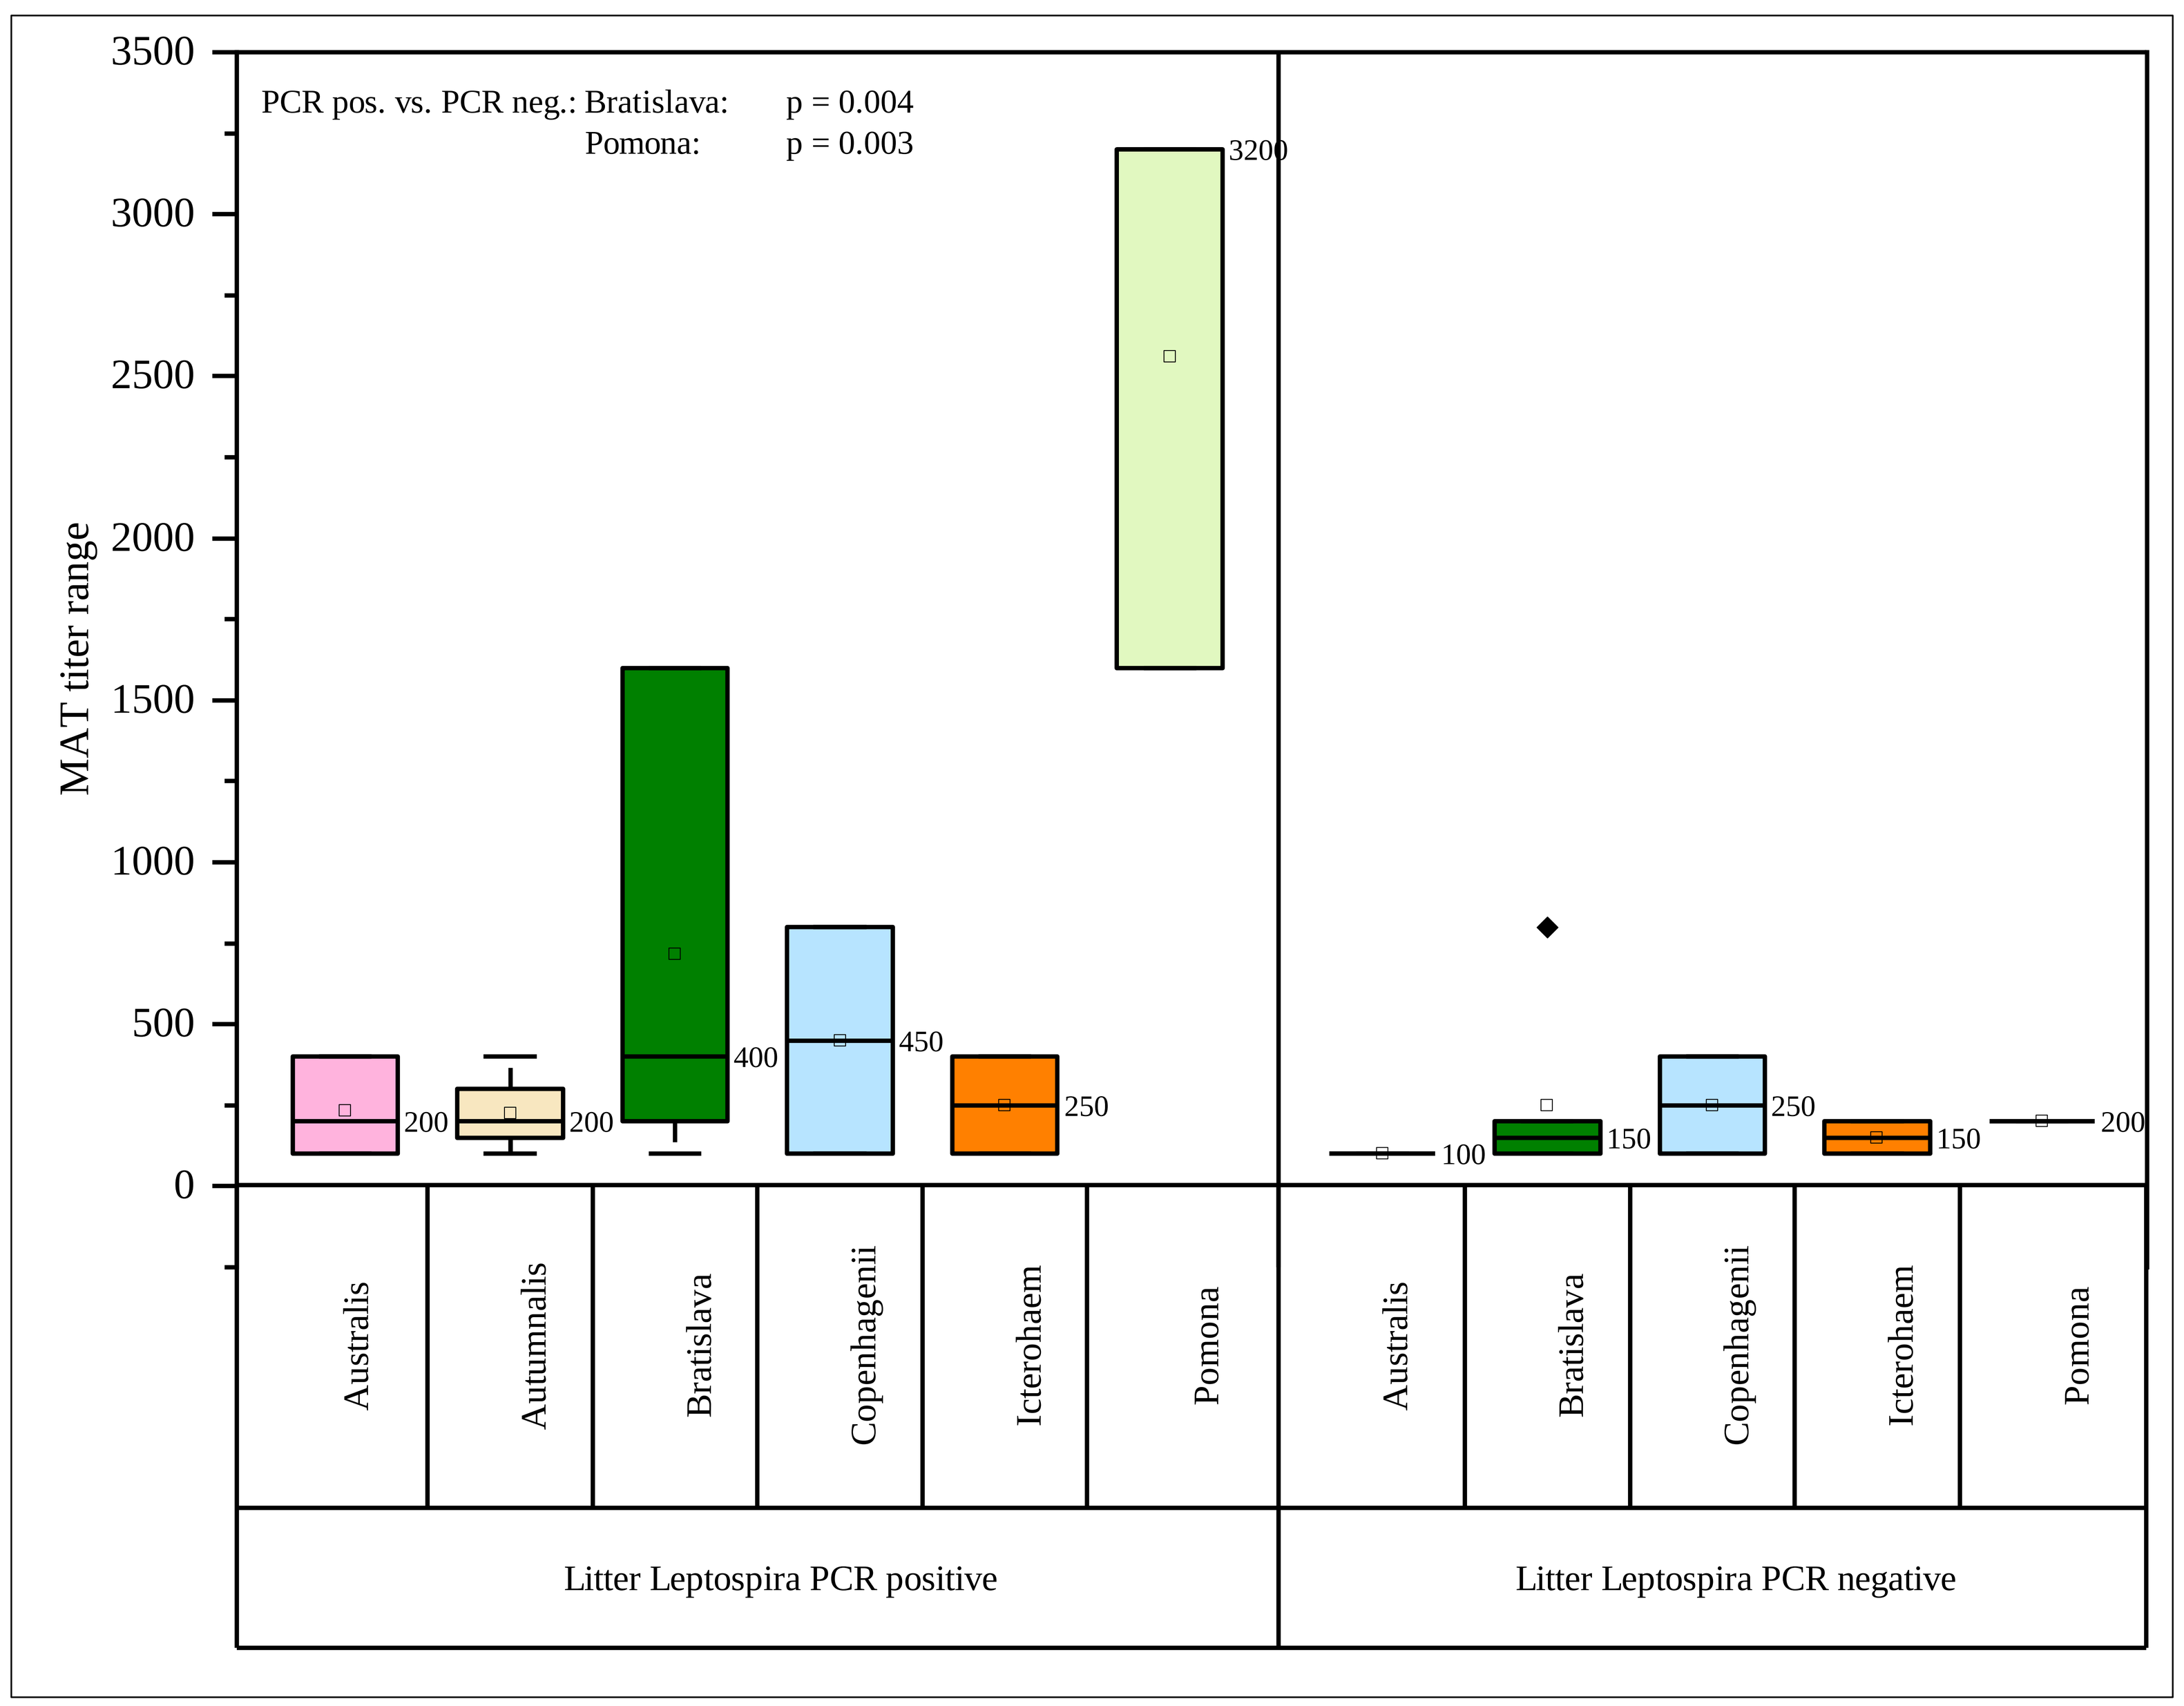

MAT titer range
3500
3000
2500
2000
1500
1000
500
0
PCR pos. vs. PCR neg.: Bratislava: p = 0.004
Pomona: p = 0.003
200
200
400
450
250
3200
100
150
250
150
200
Australis
Autumnalis
Bratislava
Copenhagenii
Icterohaem
Pomona
Litter Leptospira PCR positive
Litter Leptospira PCR negative

Figure 2: Boxplot of MAT titers in sera of sows with *Leptospira* spp. PCR positive or negative litters including median values. Only farms without *Leptospira* spp. vaccination included